Early Migraine Response by Month 1 and Clinically Meaningful Improvements in Health-Related Quality of Life (HRQoL) in Patients With Migraine in Phase 3 Trials of Eptinezumab

Stewart Tepper,[1] David Kudrow,[2] Ruslan Horblyuk,[3] Roger Cady,[4] Eric Kassel,[4] Joe Hirman[5]

[1]Geisel School of Medicine at Dartmouth, Lebanon, NH, United States; [2]California Medical Clinic for Headache, Santa Monica, CA, United States; [3]AESARA, Inc., Chapel Hill, NC; [4]Alder BioPharmaceuticals, Inc., Bothell, WA, United States; [5]Pacific Northwest Statistical Consulting, Inc., Woodinville, WA, United States

Scan here to obtain a PDF of this poster.

Copies of this poster obtained through QR code are for personal use only and may not be reproduced without written permission of the authors.

QR code

10-020

Presented at the 2019 American Academy of Neurology Annual Meeting
May 4-10, 2019
Philadelphia, PA

BACKGROUND

- Migraine is a disabling and costly neurological disorder and is estimated to affect more than 1.3 billion individuals worldwide, resulting in more than 47 million person-years lived with disability[1,2]
- Migraine, especially with increasing frequency and severity, can significantly worsen an individual's health-related quality of life (HRQOL)[3-5]
- Calcitonin gene-related peptide (CGRP) plays an important role in migraine pathophysiology[6-8]
- Eptinezumab (ALD403) is a humanized IgG1 anti-CGRP monoclonal antibody that: binds the CGRP ligand with high affinity resulting in potent and sustained inactivation of CGRP, is designed for rapid onset and durability ($t_{1/2}$ of 27 days), and is administered quarterly by intravenous (IV) infusion, allowing for 100% bioavailability with immediate C_{max} at the end of infusion[9,10]
- In phase 3 clinical trials in patients with episodic migraine (EM; NCT02559895) and chronic migraine (CM; NCT02974153), eptinezumab demonstrated significant reductions in mean monthly migraine days

OBJECTIVE

- To evaluate changes in HRQOL at Month 3 by magnitude of migraine response by Month 1 in adults with EM or CM in clinical trials of eptinezumab

METHODS

- PROMISE-1 and PROMISE-2 were phase 3, parallel-group, double-blind, randomized, placebo-controlled trials of eptinezumab for the preventive treatment of migraine
- PROMISE-1 evaluated up to 4 quarterly IV infusions of eptinezumab or placebo in patients with EM
 - After a 28-day screening period, infusions of eptinezumab or placebo were administered on Day 0 and at Weeks 12, 24, and 36, with an end-of-study visit occurring at Week 56
- PROMISE-2 evaluated up to 2 quarterly IV infusions of eptinezumab or placebo in patients with CM
 - After a 28-day screening period, infusions of eptinezumab or placebo were administered on Day 0 and at Weeks 12, with an end-of-study visit occurring at Week 32
- HRQOL was evaluated descriptively using the Short-Form Health Survey (SF-36, v2.0)—a robust measure of general health (**Table 1**)—which was administered at the baseline, Month 1, and Month 3 visits in both studies
- The SF-36 was scored using norm-based scoring, which is the standardization of mean scores and standard deviations for all SF-36 domains and components scores to a mean of 50 and standard deviations of 10, which represents the general US population[11]
 - Norm-based scoring provides a basis for comparisons across scales and more clearly reflects the impact of a disease or treatment
 - When a scale score is below 50, health status is below average, and each point is 1/10 of a standard deviation
- Clinically meaningful change in SF-36 scores is estimated between 2.5 and 5 points, which varies across disease states[12-20]
 - Here, we use a 3-point increase as the threshold for clinically meaningful change
- Subgroups were defined by ≥50%, 50%–<75%, or ≥75% migraine response by Month 1

Table 1. Descriptions of SF-36 Domain and Component Scores[11]

	Definition of Lowest Possible Score	Definition of Highest Possible Score
Domains		
Physical function (PF; 10 items)	Very limited in performing all physical activities, including bathing or dressing	Performs all types of physical activities including the most vigorous without limitations due to health
Physical role function (RP; 4 items)	Problems with work or other daily activities as a result of physical health	No problems with work or other daily activities
Bodily pain (BP; 2 items)	Very severe and extremely limiting pain	No pain or limitations due to pain
General health (GH; 5 items)	Evaluates personal health as poor and believes it is likely to get worse	Evaluates personal health as excellent
Mental health (MH; 5 items)	Feelings of nervousness and depression all of the time	Feels peaceful, happy, and calm all of the time
Emotional role function (RE; 3 items)	Problems with work or other daily activities as a result of emotional problems	No problems with work or other daily activities
Social function (SF; 2 items)	Extreme and frequent interference with normal social activities due to physical and emotional problems	Performs normal social activities without interference due to physical or emotional problems
Vitality (V; 4 items)	Feels tired and worn out all of the time	Feels full of pep and energy all of the time
Components		
Physical component summary (PCS)	Limitations in self-care, physical, social, and role activities, severe bodily pain, frequent tiredness, health rated "poor"	No physical limitations, disabilities, or decrements in well-being, high energy level, health rated "excellent"
Mental component summary (MCS)	Frequent psychological distress, social and role disability due to emotional problems, health rated "poor"	Frequent positive affect, absence of psychological distress and limitations in usual social/role activities due to emotional problems, health rated "excellent"

*Each domain score contributes to the component scores; however, the first 4 domains listed contribute the most to the PCS score and the latter 4 contribute the most to the MCS score.

CONCLUSIONS

- More than half of patients treated with eptinezumab who were ≥50% migraine responders achieved the ≥75% migraine responder threshold, with eptinezumab treatment resulting in more patients achieving either threshold vs placebo
- Patients with CM reported below normative HRQOL across all SF-36 domains and component summary scores at baseline
 - Patients achieving early and meaningful reduction in MMD frequency (≥50% migraine response) experienced clinically meaningful improvements across nearly all domains and component summary scores at Month 3
- Patients with EM reported HRQOL below normative levels in relation to Bodily Pain, Physical Role Function, and Physical Component Summary scores at baseline
 - Patients achieving early and meaningful reduction in MMD frequency (≥50% migraine response) experienced clinically meaningful improvements in Bodily Pain at Month 3, with mean increases in scores for Physical Role Function and Social Function moving patients toward normative levels
- The changes observed in SF-36 individual domains and component scores were proportional to the respective level of impairment at baseline, with eptinezumab-treated patients reaching normative or near-normative levels by Month 3 of treatment
- Treatment with eptinezumab generally resulted in greater changes from baseline in SF-36 scores across subgroups compared with placebo, indicating that eptinezumab treatment improves HRQOL beyond reducing monthly migraine days

RESULTS

- Primary and key secondary results are presented in Podium Presentation 003 (PROMISE-1) and Poster 10-006 (PROMISE-2)

Table 2. Baseline Demographics and Disease Characteristics (Safety Population)

	PROMISE-1			PROMISE-2		
	Eptinezumab 100 mg n=223	Eptinezumab 300 mg n=224	Placebo n=222	Eptinezumab 100 mg n=356	Eptinezumab 300 mg n=350	Placebo n=366
Age (years), mean (SD)	40.0 (10.66)	40.2 (11.72)	39.9 (11.67)	41.0 (11.72)	41.0 (10.36)	39.6 (11.28)
Sex, n (%)						
Male	44 (19.7%)	25 (11.2%)	36 (16.2%)	49 (13.8%)	36 (10.3%)	41 (11.2%)
Female	179 (80.3%)	199 (88.8%)	186 (83.8%)	307 (86.2%)	314 (89.7%)	325 (88.8%)
Race, n (%)						
White	196 (87.9%)	187 (83.5%)	181 (81.5%)	332 (93.3%)	322 (92.0%)	321 (87.7%)
Black or African American	17 (7.6%)	27 (12.1%)	30 (13.5%)	21 (5.9%)	23 (6.6%)	38 (10.4%)
Other*	10 (4.5%)	10 (4.4%)	11 (5.0%)	3 (0.8%)	5 (1.4%)	7 (1.9%)
BMI (kg/m^2), mean (SD)	29.4 (7.66)	28.9 (7.14)	29.6 (7.28)	26.4 (4.98)	26.3 (5.04)	27.0 (5.56)
Age at migraine diagnosis (years), mean (SD)	22.5 (10.77)	22.0 (9.87)	23.1 (10.87)	22.8 (10.64)	22.0 (9.30)	22.6 (9.98)
Duration of migraine diagnosis (years), mean (SD)	17.4 (11.18)	18.2 (11.75)	16.9 (11.23)	18.3 (12.22)	19.0 (11.50)	17.0 (11.63)
Duration of chronic migraines (years), mean (SD)	—	—	—	11.6 (11.72)	12.3 (11.15)	11.6 (10.90)
Number of migraine days, mean (SD)†	8.7 (2.85)	8.6 (2.87)	8.4 (2.68)	16.1 (4.61)	16.1 (4.77)	16.2 (4.55)
Medication-overuse headache diagnosis, n (%)§	—	—	—	139 (39.0%)	147 (42.0%)	145 (39.6%)

BMI, body mass index; SD, standard deviation; SF-36, 36-item Short-Form Health Survey (v2.0). *Other includes Asian, American Indian or Alaska Native, Native Hawaiian or Other Pacific Islander, multiple races, and other. †As reported by the eDiary in the in the 28-day screening period. §Based on 3rd edition of the International Classification of Headache Disorders (beta).

- Baseline demographics and disease characteristics were generally balanced across treatment arms and across studies (**Table 2**)
 - Patients with EM experienced ~8.6 migraine days/month, and patients with CM experienced ~16.1 migraine days/month
 - Approximately 40% of patients in PROMISE-2 had a dual diagnosis of CM and medication-overuse headache

Table 3. Summary of Migraine Responder Rates (MRRs) by Month 1

	PROMISE-1			PROMISE-2		
	Eptinezumab 100 mg n=221	Eptinezumab 300 mg n=222	Placebo n=222	Eptinezumab 100 mg n=356	Eptinezumab 300 mg n=350	Placebo n=366
≥50% MRR, n (%)	131 (59.3%)	125 (56.3%)	90 (40.5%)	194 (54.5%)	212 (60.6%)	132 (36.1%)
50%–<75% MRR, n (%)	63 (28.5%)	55 (24.8%)	45 (20.3%)	84 (23.6%)	83 (23.7%)	75 (20.5%)
≥75% MRR, n (%)	68 (30.8%)	70 (31.5%)	45 (20.3%)	110 (30.9%)	129 (36.9%)	57 (15.6%)

- In patients achieving ≥50% migraine response at Month 1, more eptinezumab-treated patients were ≥75% migraine responders than 50%–<75% migraine responders; in contrast, an equal proportion of placebo-treated patients were ≥75% and 50%–<75% migraine responders in PROMISE-1 and less were ≥75% than 50%–<75% migraine responders in PROMISE-2 (**Table 3**)

Figure 1. Baseline SF-36 Scores in PROMISE-1 (Total Population)

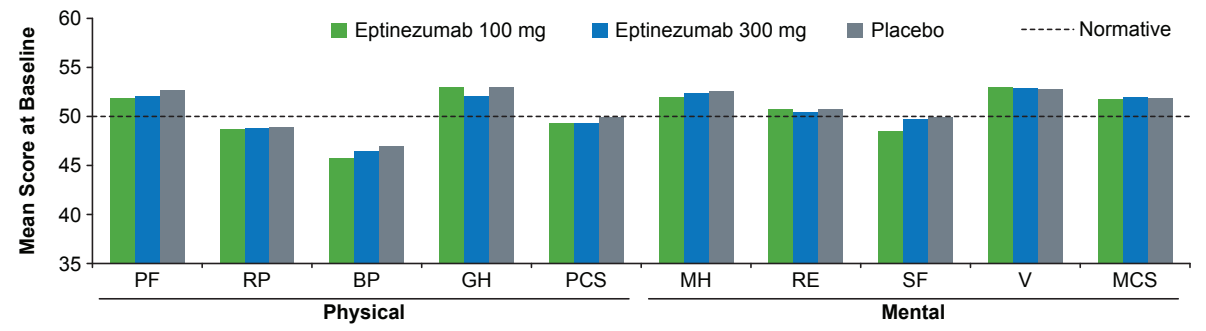
Eptinezumab 100 mg
Eptinezumab 300 mg
Placebo
Normative
Mean Score at Baseline
60
55
50
45
40
35
PF
RP
BP
GH
PCS
MH
RE
SF
V
MCS
Physical
Mental

See **Table 1** for SF-36 acronyms.

- At PROMISE-1 baseline, there were noticeable impairments in the Bodily Pain and Physical Role Function domains of the SF-36, as well as a score below the normative line HRQOL in the Social Function domain, across patients with EM (**Figure 1**)

Figure 2. PROMISE-1 Change From Baseline to Month 3 in Most Impacted SF-36 Scores by Migraine Response at Month 1

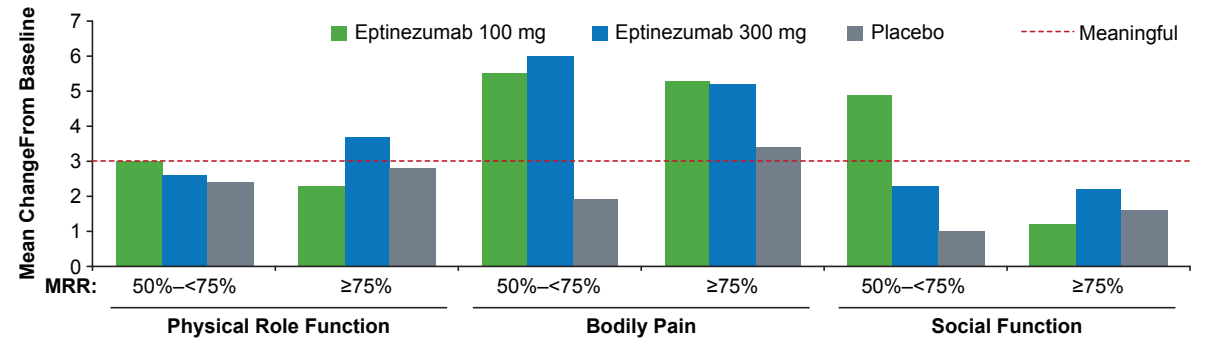
Eptinezumab 100 mg
Eptinezumab 300 mg
Placebo
Meaningful
Mean ChangeFrom Baseline
7
6
5
4
3
2
1
0
MRR:
50%–<75%
≥75%
50%–<75%
≥75%
50%–<75%
≥75%
Physical Role Function
Bodily Pain
Social Function

MRR, migraine responder rate.

- By Month 3, clinically meaningful improvements were observed for Bodily Pain in both 50%–<75% and ≥75% migraine responders, for Physical Role Function in ≥75% migraine responders, and for Social Function in 50%–<75% migraine responders (**Figure 2**)
- For scores that were normative at baseline, patients maintained at the normative line or incremental improvements were observed
- Changes in SF-36 scores were generally greater with eptinezumab treatment than placebo

Figure 3. Baseline SF-36 Scores in PROMISE-2 (Total Population)

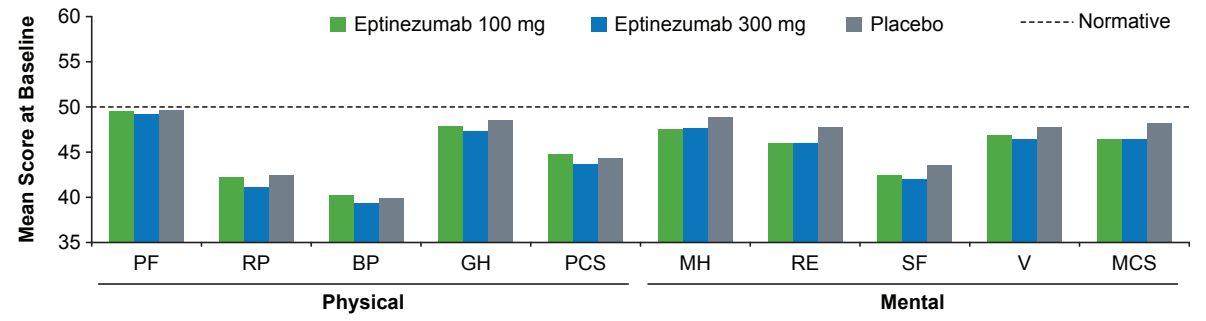
Eptinezumab 100 mg
Eptinezumab 300 mg
Placebo
Normative
Mean Score at Baseline
60
55
50
45
40
35
PF
RP
BP
GH
PCS
MH
RE
SF
V
MCS
Physical
Mental

See **Table 1** for SF-36 acronyms.

- At PROMISE-2 baseline, all domains and component summary scores were below normative levels in patients with CM, with the largest impairments observed for Bodily Pain, Physical Role Function, and Social Functioning (**Figure 3**)

Figure 4. PROMISE-2 Change From Baseline to Month 3 Physical SF-36 Scores by Migraine Response at Month 1

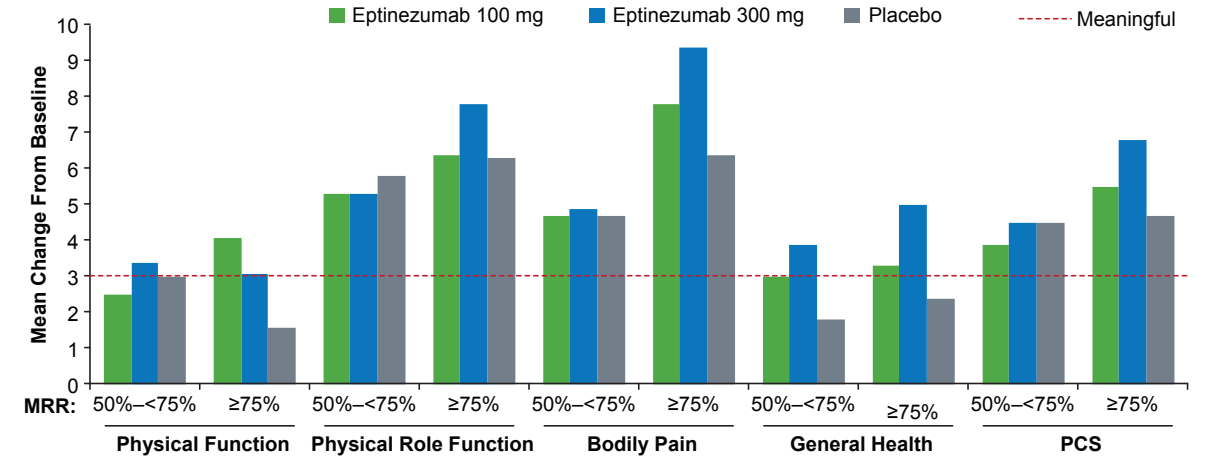
Eptinezumab 100 mg
Eptinezumab 300 mg
Placebo
Meaningful
Mean Change From Baseline
10
9
8
7
6
5
4
3
2
1
0
MRR:
50%–<75%
≥75%
Physical Function
Physical Role Function
Bodily Pain
General Health
PCS

Figure 5. PROMISE-2 Change From Baseline to Month 3 in Mental SF-36 Scores by Migraine Response at Month 1

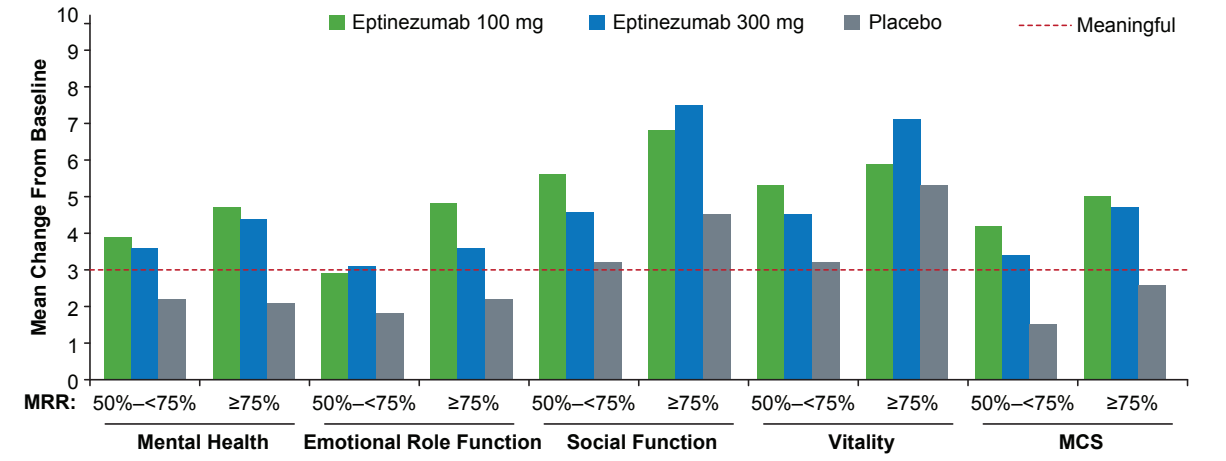
Eptinezumab 100 mg
Eptinezumab 300 mg
Placebo
Meaningful
Mean Change From Baseline
10
9
8
7
6
5
4
3
2
1
0
MRR:
50%–<75%
≥75%
Mental Health
Emotional Role Function
Social Function
Vitality
MCS

MRR, migraine responder rate.

- **Figure 4** depicts change from baseline to Month 3 in SF-36 scores related to physical aspects of HRQOL; **Figure 5** depicts the mental aspects
- By Month 3, clinically meaningful improvements were observed across almost all SF-36 domains and component scores for eptinezumab-treated patients achieving early (Month 1) migraine response
 - Larger increases in scores were observed for patients experiencing greater reduction on MMDs
- Improvements in SF-36 scores were greater with eptinezumab than with placebo for all mental domains, most physical domains, and both component summary scores
- Placebo-treated patients achieving ≥75% migraine response by Month 1 experienced clinically meaningful improvements across half of the domains and PCS score

Table 4. Mean (SD) Change From Baseline to Month 3 in SF-36 Scores by Migraine Response at Month 1

	PROMISE-1			PROMISE-2		
	Eptinezumab 100 mg	Eptinezumab 300 mg	Placebo	Eptinezumab 100 mg	Eptinezumab 300 mg	Placebo
≥50% MRR						
PF	*0.6*	*1.2*	*0.8*	**3.4**	**3.3**	2.4
RP	2.6	**3.2**	2.6	**6.0**	**6.9**	**6.0**
BP	5.4	**5.5**	2.7	**6.5**	**7.7**	**5.4**
GH	*0.8*	*1.6*	*1.6*	**3.2**	**4.6**	2.1
MH	*1.3*	*1.1*	*1.0*	**4.3**	**4.1**	2.1
RE	0.7	*1.3*	*1.9*	**4.0**	**3.4**	2.0
SF	**3.0**	2.2	1.3	**6.3**	**6.4**	**3.7**
V	*1.8*	2.2	*2.1*	**5.7**	**6.1**	**4.1**
PCS	2.7	**3.3**	2.0	**4.8**	**5.9**	**4.6**
MCS	1.1	1.0	1.3	**4.6**	**4.2**	2.0
50%–<75% MRR						
PF	*0.2*	*1.7*	*0.4*	2.5	**3.4**	**3.0**
RP	**3.0**	*2.6*	2.4	**5.3**	**5.3**	**5.8**
BP	**5.5**	**6.0**	1.9	**4.7**	**4.9**	**4.7**
GH	*0.2*	*1.8*	*1.6*	**3.0**	**3.9**	1.8
MH	*0.1*	*0.5*	*-0.3*	**3.9**	**3.6**	2.2
RE	*0.1*	*1.0*	*2.1*	2.9	**3.1**	1.8
SF	**4.9**	2.3	1.0	**5.6**	**4.6**	**3.2**
V	*1.4*	2.3	*2.5*	**5.3**	**4.5**	**3.2**
PCS	2.9	**3.7**	1.8	**3.9**	**4.5**	**4.5**
MCS	0.8	0.5	1.0	**4.2**	**3.4**	1.5
≥75% MRR						
PF	*1.0*	*0.7*	*1.3*	**4.1**	**3.1**	1.6
RP	*2.3*	**3.7**	*2.8*	**6.4**	**7.8**	**6.3**
BP	**5.3**	**5.2**	**3.4**	**7.8**	**9.4**	**6.4**
GH	*1.3*	*1.5*	*1.6*	**3.3**	**5.0**	2.4
MH	*2.3*	*1.5*	*2.3*	**4.7**	**4.4**	2.1
RE	*1.2*	*1.6*	*1.7*	**4.8**	**3.6**	2.2
SF	1.2	*2.2*	1.6	**6.8**	**7.5**	**4.5**
V	*2.1*	*2.2*	*1.8*	**5.9**	**7.1**	**5.3**
PCS	2.5	**3.0**	2.2	**5.5**	**6.8**	**4.7**
MCS	1.4	1.4	1.7	**5.0**	**4.7**	2.6

MRR, migraine responder rate; SD, standard deviation. See **Table 1** for SF-36 acronyms.

- **Table 4** displays the mean changes from baseline across each of the SF-36 domain and component summary scores by migraine responder thresholds.
 - Bold values indicate that the changes were considered clinically meaningful (ie, change of ≥3)
 - Italicized values indicate that the baseline scores were in the normative range (ie, score ≥50)

References: 1. Lipton RB, Silberstein SD. *Headache*. 2015. doi: 10.1111/head.12505_2. **2.** GBD 2017 Disease and Injury Incidence and Prevalence Collaborators. *Lancet*. 2017. doi: 10.1016/S0140-6736(18)32279-7. **3.** Cho SJ, et al. *Curr Pain Headache Rep*. 2016. doi: 10.1007/s11916-015-0534-8. **4.** Blumenfeld AM, et al. *Cephalalgia*. 2011. doi: 10.1177/0333102410381145. **5.** Freitag FG. *Clin Ther*. 2007. doi: 10.1016/j.clinthera.2007.05.008. **6.** Lassen LH, et al. *Cephalalgia*. 2002. doi: 10.1046/j.1468-2982.2002.00310.x. **7.** Hansen JM, et al. *Cephalalgia*. 2010. doi: 10.1177/0333102410368444. **8.** Ho TW, et al. *Nat Rev Neurol*. 2010. doi: 10.1038/nrneurol.2010.127. **9.** Baker B, et al. [abstract]. *Neurology*. 2017;88(16 Suppl): P2.155. **10.** Dodick DW, et al. *Lancet Neurol*. 2014. doi: 10.1016/S1474-4422(14)70209-1. **11.** Ware JE Jr. *Spine (Phila Pa 1976)*. 2000;25(24):3130-9. **12.** Angst F, et al. *Arthritis Care Res*. 2001. doi: 10.1002/1529-0131(200108)45:4%3C384::AID-ART352%3E3.0.CO;2-0. **13.** Ward MM, et al. *Arthritis Care Res (Hoboken)*. 2014. doi: 10.1002/acr.22392. **14.** Kosinski M, et al. *Arthritis Rheum*. 2000. doi: 10.1002/1529-0131(200007)43:7<1478::AID-ANR10>3.0.CO;2-M. **15.** Badhiwala JH, et al. *Spine (Phila Pa 1976)*. 2018. doi: 10.1097/BRS.0000000000002684. **16.** Carreon LY, et al. *Spine J*. 2010. doi: 10.1016/j.spinee.2010.02.007. **17.** Yuksel S, et al. *Spine J*. 2019. doi: 10.1016/j.spinee.2018.07.005. **18.** Copay AG, et al. *Spine J*. 2008. doi: 10.1016/j.spinee.2007.11.006. **19.** Carlson et al. *Otolaryngol Head Neck Surg*. 2015. doi: 10.3171/2014.11.JNS14594/. **20.** Swigris et al. *Respir Med*. 2010. doi: 10.1016/j.rmed.2009.09.006.

Acknowledgments: The authors thank Nicole Coolbaugh of The Medicine Group (New Hope, PA, USA) for providing medical writing support, which was funded by Alder BioPharmaceuticals, Inc. (Bothell, WA, USA) in accordance with Good Publication Practice guidelines.

Disclosures: S. Tepper: *Consultant and/or scientific advisor:* Acorda, Alder BioPharmaceuticals, Alexsa, Allergan, Alphasights, Amgen, ATI, Axsome Therapeutics, Cefaly, Charleston Labs, DeepBench, Dr. Reddy's, ElectroCore, Eli Lilly, eNeura, GLG, Guidepoint Global, Magellan Rx Management, Neurolief, Nordic BioTech, Pfizer, Scion Neurostim, Slingshot Insights, Supernus, Teva, Zosano; *Editor and/or advising editor:* American Headache Society, Headache Currents, Headache, Wiley Blackwell; *Research support:* Alder BioPharmaceuticals, Allergan, Amgen, ATI, Dr. Reddy's Laboratories, ElectroCore, eNeura, Scion Neurostim, Teva, Zosano.: **D. Kudrow:** *Consultant and/or scientific advisor:* Alder BioPharmaceuticals, Amgen, Biohaven, Eli Lilly, Promius; *Research support:* Alder BioPharmaceuticals, Allergan, Amgen, Biohaven, Eli Lilly, Genentech, Roche, Teva, Viromed.: **R. Horblyuk:** *Contracted service provider:* Alder BioPharmaceuticals. **R. Cady:** *Full-time employee:* Alder BioPharmaceuticals; *Stockholder:* Alder BioPharmaceuticals. **E. Kassel:** *Full-time employee:* Alder BioPharmaceuticals; *Stockholder:* Alder BioPharmaceuticals. **J. Hirman:** *Contracted service provider:* Alder BioPharmaceuticals.

This study was funded by Alder BioPharmaceuticals, Inc.

Early Migraine Response by Month 1 and Clinically Meaningful Improvements in Health-Related Quality of Life (HRQoL) in Patients With Migraine in Phase 3 Trials of Eptinezumab

Stewart Tepper,[1] David Kudrow,[2] Ruslan Horblyuk,[3] Roger Cady,[4] Eric Kassel,[4] Joe Hirman[5]

[1]Geisel School of Medicine at Dartmouth, Lebanon, NH, United States; [2]California Medical Clinic for Headache, Santa Monica, CA, United States; [3]AESARA, Inc., Chapel Hill, NC; [4]Alder BioPharmaceuticals, Inc., Bothell, WA, United States; [5]Pacific Northwest Statistical Consulting, Inc., Woodinville, WA, United States

Scan here to obtain a PDF of this poster.

Copies of this poster obtained through QR code are for personal use only and may not be reproduced without written permission of the authors.

QR code

10-020

Presented at the 2019 American Academy of Neurology Annual Meeting May 4-10, 2019 Philadelphia, PA

BACKGROUND

- Migraine is a disabling and costly neurological disorder and is estimated to affect more than 1.3 billion individuals worldwide, resulting in more than 47 million person-years lived with disability[1,2]
- Migraine, especially with increasing frequency and severity, can significantly worsen an individual's health-related quality of life (HRQOL)[3-5]
- Calcitonin gene-related peptide (CGRP) plays an important role in migraine pathophysiology[6-8]
- Eptinezumab (ALD403) is a humanized IgG1 anti-CGRP monoclonal antibody that: binds the CGRP ligand with high affinity resulting in potent and sustained inactivation of CGRP, is designed for rapid onset and durability ($t_{1/2}$ of 27 days), and is administered quarterly by intravenous (IV) infusion, allowing for 100% bioavailability with immediate C_{max} at the end of infusion[9,10]
- In phase 3 clinical trials in patients with episodic migraine (EM; NCT02559895) and chronic migraine (CM; NCT02974153), eptinezumab demonstrated significant reductions in mean monthly migraine days

OBJECTIVE

- To evaluate changes in HRQOL at Month 3 by magnitude of migraine response by Month 1 in adults with EM or CM in clinical trials of eptinezumab

METHODS

- PROMISE-1 and PROMISE-2 were phase 3, parallel-group, double-blind, randomized, placebo-controlled trials of eptinezumab for the preventive treatment of migraine
- PROMISE-1 evaluated up to 4 quarterly IV infusions of eptinezumab or placebo in patients with EM
 - After a 28-day screening period, infusions of eptinezumab or placebo were administered on Day 0 and at Weeks 12, 24, and 36, with an end-of-study visit occurring at Week 56
- PROMISE-2 evaluated up to 2 quarterly IV infusions of eptinezumab or placebo in patients with CM
 - After a 28-day screening period, infusions of eptinezumab or placebo were administered on Day 0 and at Weeks 12, with an end-of-study visit occurring at Week 32
- HRQOL was evaluated descriptively using the Short-Form Health Survey (SF-36, v2.0)—a robust measure of general health (**Table 1**)—which was administered at the baseline, Month 1, and Month 3 visits in both studies
- The SF-36 was scored using norm-based scoring, which is the standardization of mean scores and standard deviations for all SF-36 domains and components scores to a mean of 50 and standard deviations of 10, which represents the general US population[11]
 - Norm-based scoring provides a basis for comparisons across scales and more clearly reflects the impact of a disease or treatment
 - When a scale score is below 50, health status is below average, and each point is 1/10 of a standard deviation
- Clinically meaningful change in SF-36 scores is estimated between 2.5 and 5 points, which varies across disease states[12-20]
 - Here, we use a 3-point increase as the threshold for clinically meaningful change
- Subgroups were defined by ≥50%, 50%–<75%, or ≥75% migraine response by Month 1

Table 1. Descriptions of SF-36 Domain and Component Scores[11]

	Definition of Lowest Possible Score	Definition of Highest Possible Score
Domains		
Physical function (PF; 10 items)	Very limited in performing all physical activities, including bathing or dressing	Performs all types of physical activities including the most vigorous without limitations due to health
Physical role function (RP; 4 items)	Problems with work or other daily activities as a result of physical health	No problems with work or other daily activities
Bodily pain (BP; 2 items)	Very severe and extremely limiting pain	No pain or limitations due to pain
General health (GH; 5 items)	Evaluates personal health as poor and believes it is likely to get worse	Evaluates personal health as excellent
Mental health (MH; 5 items)	Feelings of nervousness and depression all of the time	Feels peaceful, happy, and calm all of the time
Emotional role function (RE; 3 items)	Problems with work or other daily activities as a result of emotional problems	No problems with work or other daily activities
Social function (SF; 2 items)	Extreme and frequent interference with normal social activities due to physical and emotional problems	Performs normal social activities without interference due to physical or emotional problems
Vitality (V; 4 items)	Feels tired and worn out all of the time	Feels full of pep and energy all of the time
Components		
Physical component summary (PCS)	Limitations in self-care, physical, social, and role activities, severe bodily pain, frequent tiredness, health rated "poor"	No physical limitations, disabilities, or decrements in well-being, high energy level, health rated "excellent"
Mental component summary (MCS)	Frequent psychological distress, social and role disability due to emotional problems, health rated "poor"	Frequent positive affect, absence of psychological distress and limitations in usual social/role activities due to emotional problems, health rated "excellent"

*Each domain score contributes to the component scores; however, the first 4 domains listed contribute the most to the PCS score and the latter 4 contribute the most to the MCS score.

CONCLUSIONS

- More than half of patients treated with eptinezumab who were ≥50% migraine responders achieved the ≥75% migraine responder threshold, with eptinezumab treatment resulting in more patients achieving either threshold vs placebo
- Patients with CM reported below normative HRQOL across all SF-36 domains and component summary scores at baseline
 - Patients achieving early and meaningful reduction in MMD frequency (≥50% migraine response) experienced clinically meaningful improvements across nearly all domains and component summary scores at Month 3
- Patients with EM reported HRQOL below normative levels in relation to Bodily Pain, Physical Role Function, and Physical Component Summary scores at baseline
 - Patients achieving early and meaningful reduction in MMD frequency (≥50% migraine response) experienced clinically meaningful improvements in Bodily Pain at Month 3, with mean increases in scores for Physical Role Function and Social Function moving patients toward normative levels
- The changes observed in SF-36 individual domains and component scores were proportional to the respective level of impairment at baseline, with eptinezumab-treated patients reaching normative or near-normative levels by Month 3 of treatment
- Treatment with eptinezumab generally resulted in greater changes from baseline in SF-36 scores across subgroups compared with placebo, indicating that eptinezumab treatment improves HRQOL beyond reducing monthly migraine days

- Primary and key secondary results are presented in Podium Presentation 003 (PROMISE-1) and Poster 10-006 (PROMISE-2)

Table 2. Baseline Demographics and Disease Characteristics (Safety Population)

	PROMISE-1			PROMISE-2		
	Eptinezumab 100 mg n=223	Eptinezumab 300 mg n=224	Placebo n=222	Eptinezumab 100 mg n=356	Eptinezumab 300 mg n=350	Placebo n=366
Age (years), mean (SD)	40.0 (10.66)	40.2 (11.72)	39.9 (11.67)	41.0 (11.72)	41.0 (10.36)	39.6 (11.28)
Sex, n (%)						
Male	44 (19.7%)	25 (11.2%)	36 (16.2%)	49 (13.8%)	36 (10.3%)	41 (11.2%)
Female	179 (80.3%)	199 (88.8%)	186 (83.8%)	307 (86.2%)	314 (89.7%)	325 (88.8%)
Race, n (%)						
White	196 (87.9%)	187 (83.5%)	181 (81.5%)	332 (93.3%)	322 (92.0%)	321 (87.7%)
Black or African American	17 (7.6%)	27 (12.1%)	30 (13.5%)	21 (5.9%)	23 (6.6%)	38 (10.4%)
Other*	10 (4.5%)	10 (4.4%)	11 (5.0%)	3 (0.8%)	5 (1.4%)	7 (1.9%)
BMI (kg/m^2), mean (SD)	29.4 (7.66)	28.9 (7.14)	29.6 (7.28)	26.4 (4.98)	26.3 (5.04)	27.0 (5.56)
Age at migraine diagnosis (years), mean (SD)	22.5 (10.77)	22.0 (9.87)	23.1 (10.87)	22.8 (10.64)	22.0 (9.30)	22.6 (9.98)
Duration of migraine diagnosis (years), mean (SD)	17.4 (11.18)	18.2 (11.75)	16.9 (11.23)	18.3 (12.22)	19.0 (11.50)	17.0 (11.63)
Duration of chronic migraines (years), mean (SD)	—	—	—	11.6 (11.72)	12.3 (11.15)	11.6 (10.90)
Number of migraine days, mean (SD)†	8.7 (2.85)	8.6 (2.87)	8.4 (2.68)	16.1 (4.61)	16.1 (4.77)	16.2 (4.55)
Medication-overuse headache diagnosis, n (%)§	—	—	—	139 (39.0%)	147 (42.0%)	145 (39.6%)

BMI, body mass index; SD, standard deviation; SF-36, 36-item Short-Form Health Survey (v2.0). *Other includes Asian, American Indian or Alaska Native, Native Hawaiian or Other Pacific Islander, multiple races, and other. †As reported by the eDiary in the in the 28-day screening period. §Based on 3rd edition of the International Classification of Headache Disorders (beta).

- Baseline demographics and disease characteristics were generally balanced across treatment arms and across studies (Table 2)
 - Patients with EM experienced ~8.6 migraine days/month, and patients with CM experienced ~16.1 migraine days/month
 - Approximately 40% of patients in PROMISE-2 had a dual diagnosis of CM and medication-overuse headache

Table 3. Summary of Migraine Responder Rates (MRRs) by Month 1

	PROMISE-1			PROMISE-2		
	Eptinezumab 100 mg n=221	Eptinezumab 300 mg n=222	Placebo n=222	Eptinezumab 100 mg n=356	Eptinezumab 300 mg n=350	Placebo n=366
≥50% MRR, n (%)	131 (59.3%)	125 (56.3%)	90 (40.5%)	194 (54.5%)	212 (60.6%)	132 (36.1%)
50%–<75% MRR, n (%)	63 (28.5%)	55 (24.8%)	45 (20.3%)	84 (23.6%)	83 (23.7%)	75 (20.5%)
≥75% MRR, n (%)	68 (30.8%)	70 (31.5%)	45 (20.3%)	110 (30.9%)	129 (36.9%)	57 (15.6%)

- In patients achieving ≥50% migraine response at Month 1, more eptinezumab-treated patients were ≥75% migraine responders than 50%–<75% migraine responders; in contrast, an equal proportion of placebo-treated patients were ≥75% and 50%–<75% migraine responders in PROMISE-1 and less were ≥75% than 50%–<75% migraine responders in PROMISE-2 (Table 3)

Figure 1. Baseline SF-36 Scores in PROMISE-1 (Total Population)

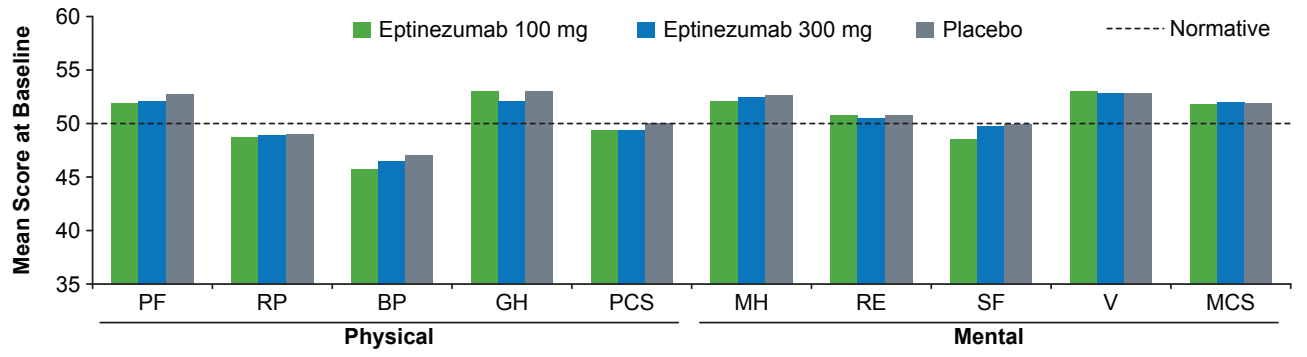

Eptinezumab 100 mg
Eptinezumab 300 mg
Placebo
Normative
Mean Score at Baseline
PF RP BP GH PCS MH RE SF V MCS
Physical
Mental

See Table 1 for SF-36 acronyms.

- At PROMISE-1 baseline, there were noticeable impairments in the Bodily Pain and Physical Role Function domains of the SF-36, as well as a score below the normative line HRQOL in the Social Function domain, across patients with EM (Figure 1)

Figure 2. PROMISE-1 Change From Baseline to Month 3 in Most Impacted SF-36 Scores by Migraine Response at Month 1

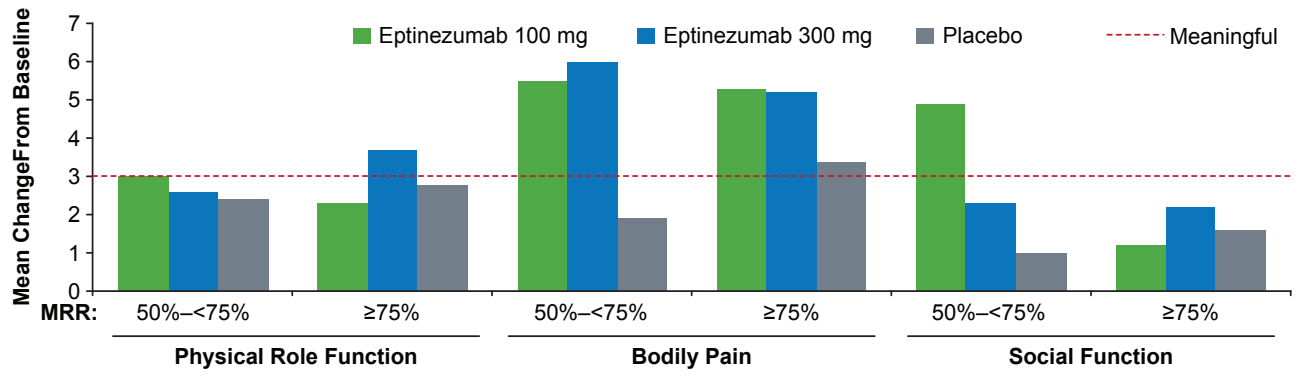

Eptinezumab 100 mg
Eptinezumab 300 mg
Placebo
Meaningful
Mean ChangeFrom Baseline
MRR:
50%–<75% ≥75% 50%–<75% ≥75% 50%–<75% ≥75%
Physical Role Function
Bodily Pain
Social Function

MRR, migraine responder rate.

- By Month 3, clinically meaningful improvements were observed for Bodily Pain in both 50%–<75% and ≥75% migraine responders, for Physical Role Function in ≥75% migraine responders, and for Social Function in 50%–<75% migraine responders (Figure 2)
- For scores that were normative at baseline, patients maintained at the normative line or incremental improvements were observed
- Changes in SF-36 scores were generally greater with eptinezumab treatment than placebo

Figure 3. Baseline SF-36 Scores in PROMISE-2 (Total Population)

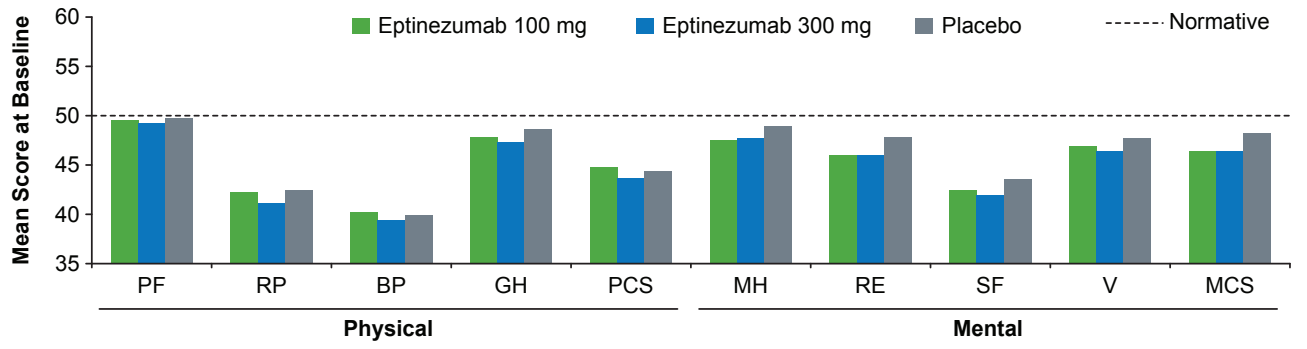

Eptinezumab 100 mg
Eptinezumab 300 mg
Placebo
Normative
Mean Score at Baseline
PF RP BP GH PCS MH RE SF V MCS
Physical
Mental

See Table 1 for SF-36 acronyms.

- At PROMISE-2 baseline, all domains and component summary scores were below normative levels in patients with CM, with the largest impairments observed for Bodily Pain, Physical Role Function, and Social Functioning (Figure 3)

Figure 4. PROMISE-2 Change From Baseline to Month 3 Physical SF-36 Scores by Migraine Response at Month 1

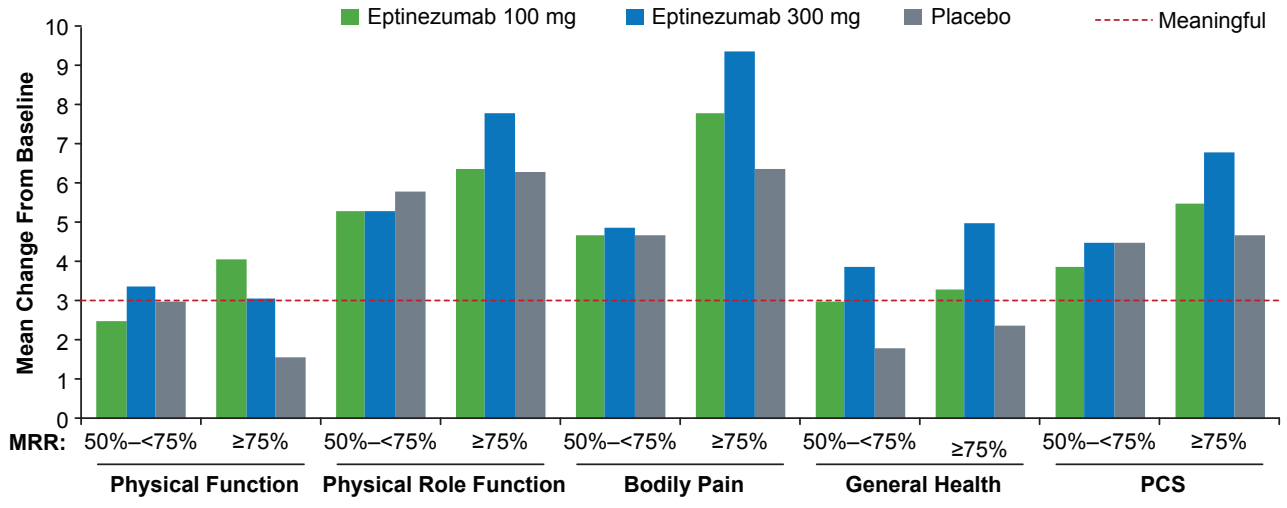

Eptinezumab 100 mg
Eptinezumab 300 mg
Placebo
Meaningful
Mean Change From Baseline
MRR:
50%–<75% ≥75% 50%–<75% ≥75% 50%–<75% ≥75% 50%–<75% ≥75% 50%–<75% ≥75%
Physical Function
Physical Role Function
Bodily Pain
General Health
PCS

Figure 5. PROMISE-2 Change From Baseline to Month 3 in Mental SF-36 Scores by Migraine Response at Month 1

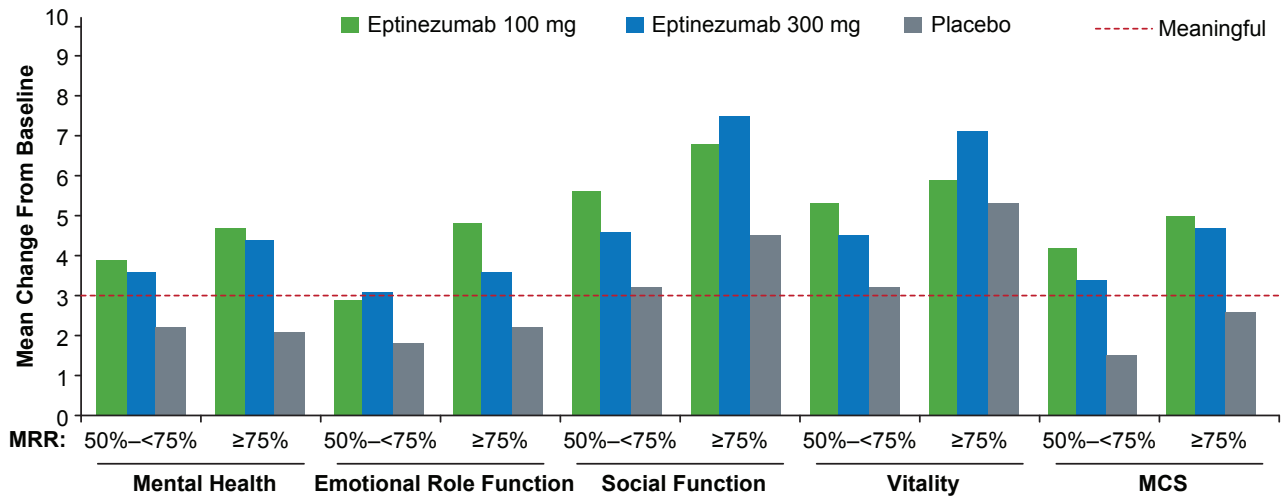

Eptinezumab 100 mg
Eptinezumab 300 mg
Placebo
Meaningful
Mean Change From Baseline
MRR:
50%–<75% ≥75% 50%–<75% ≥75% 50%–<75% ≥75% 50%–<75% ≥75% 50%–<75% ≥75%
Mental Health
Emotional Role Function
Social Function
Vitality
MCS

MRR, migraine responder rate.

- Figure 4 depicts change from baseline to Month 3 in SF-36 scores related to physical aspects of HRQOL; Figure 5 depicts the mental aspects
- By Month 3, clinically meaningful improvements were observed across almost all SF-36 domains and component scores for eptinezumab-treated patients achieving early (Month 1) migraine response
 - Larger increases in scores were observed for patients experiencing greater reduction on MMDs
- Improvements in SF-36 scores were greater with eptinezumab than with placebo for all mental domains, most physical domains, and both component summary scores
- Placebo-treated patients achieving ≥75% migraine response by Month 1 experienced clinically meaningful improvements across half of the domains and PCS score

Table 4. Mean (SD) Change From Baseline to Month 3 in SF-36 Scores by Migraine Response at Month 1

	PROMISE-1			PROMISE-2		
	Eptinezumab 100 mg	Eptinezumab 300 mg	Placebo	Eptinezumab 100 mg	Eptinezumab 300 mg	Placebo
≥50% MRR						
PF	*0.6*	*1.2*	*0.8*	**3.4**	**3.3**	2.4
RP	2.6	**3.2**	2.6	**6.0**	**6.9**	**6.0**
BP	5.4	**5.5**	2.7	**6.5**	**7.7**	**5.4**
GH	*0.8*	*1.6*	*1.6*	**3.2**	**4.6**	2.1
MH	*1.3*	*1.1*	*1.0*	**4.3**	**4.1**	2.1
RE	0.7	*1.3*	*1.9*	**4.0**	**3.4**	2.0
SF	**3.0**	2.2	1.3	**6.3**	**6.4**	**3.7**
V	*1.8*	2.2	*2.1*	**5.7**	**6.1**	**4.1**
PCS	2.7	**3.3**	2.0	**4.8**	**5.9**	**4.6**
MCS	1.1	1.0	1.3	**4.6**	**4.2**	2.0
50%–<75% MRR						
PF	*0.2*	*1.7*	*0.4*	2.5	**3.4**	**3.0**
RP	**3.0**	*2.6*	2.4	**5.3**	**5.3**	**5.8**
BP	**5.5**	**6.0**	1.9	**4.7**	**4.9**	**4.7**
GH	*0.2*	*1.8*	*1.6*	**3.0**	**3.9**	1.8
MH	*0.1*	*0.5*	*-0.3*	**3.9**	**3.6**	2.2
RE	*0.1*	*1.0*	*2.1*	2.9	**3.1**	1.8
SF	**4.9**	2.3	1.0	**5.6**	**4.6**	**3.2**
V	*1.4*	2.3	*2.5*	**5.3**	**4.5**	**3.2**
PCS	2.9	**3.7**	1.8	**3.9**	**4.5**	**4.5**
MCS	0.8	0.5	1.0	**4.2**	**3.4**	1.5
≥75% MRR						
PF	*1.0*	*0.7*	*1.3*	**4.1**	**3.1**	1.6
RP	*2.3*	**3.7**	*2.8*	**6.4**	**7.8**	**6.3**
BP	**5.3**	**5.2**	**3.4**	**7.8**	**9.4**	**6.4**
GH	*1.3*	*1.5*	*1.6*	**3.3**	**5.0**	2.4
MH	*2.3*	*1.5*	*2.3*	**4.7**	**4.4**	2.1
RE	*1.2*	*1.6*	*1.7*	**4.8**	**3.6**	2.2
SF	1.2	*2.2*	1.6	**6.8**	**7.5**	**4.5**
V	*2.1*	*2.2*	*1.8*	**5.9**	**7.1**	**5.3**
PCS	2.5	**3.0**	2.2	**5.5**	**6.8**	**4.7**
MCS	1.4	1.4	1.7	**5.0**	**4.7**	2.6

MRR, migraine responder rate; SD, standard deviation. See Table 1 for SF-36 acronyms.

- Table 4 displays the mean changes from baseline across each of the SF-36 domain and component summary scores by migraine responder thresholds.
 - Bold values indicate that the changes were considered clinically meaningful (ie, change of ≥3)
 - Italicized values indicate that the baseline scores were in the normative range (ie, score ≥50)

References: 1. Lipton RB, Silberstein SD. *Headache*. 2015. doi: 10.1111/head.12505_2. **2.** GBD 2017 Disease and Injury Incidence and Prevalence Collaborators. *Lancet*. 2017. doi: 10.1016/S0140-6736(18)32279-7. **3.** Cho SJ, et al. *Curr Pain Headache Rep*. 2016. doi: 10.1007/s11916-015-0534-8. **4.** Blumenfeld AM, et al. *Cephalalgia*. 2011. doi: 10.1177/0333102410381145. **5.** Freitag FG. *Clin Ther*. 2007. doi: 10.1016/j.clinthera.2007.05.008. **6.** Lassen LH, et al. *Cephalalgia*. 2002. doi: 10.1046/j.1468-2982.2002.00310.x. **7.** Hansen JM, et al. *Cephalalgia*. 2010. doi: 10.1177/0333102410368444. **8.** Ho TW, et al. *Nat Rev Neurol*. 2010. doi: 10.1038/nrneurol.2010.127. **9.** Baker B, et al. [abstract]. *Neurology*. 2017;88(16 Suppl): P2.155. **10.** Dodick DW, et al. *Lancet Neurol*. 2014. doi: 10.1016/S1474-4422(14)70209-1. **11.** Ware JE Jr. *Spine (Phila Pa 1976)*. 2000;25(24):3130-9. **12.** Angst F, et al. *Arthritis Care Res*. 2001. doi: 10.1002/1529-0131(200108)45:4%3C384::AID-ART352%3E3.0.CO;2-0. **13.** Ward MM, et al. *Arthritis Care Res (Hoboken)*. 2014. doi: 10.1002/acr.22392. **14.** Kosinski M, et al. *Arthritis Rheum*. 2000. doi: 10.1002/1529-0131(200007)43:7<1478::AID-ANR10>3.0.CO;2-M. **15.** Badhiwala JH, et al. *Spine (Phila Pa 1976)*. 2018. doi: 10.1097/BRS.0000000000002684. **16.** Carreon LY, et al. *Spine J*. 2010. doi: 10.1016/j.spinee.2010.02.007. **17.** Yuksel S, et al. *Spine J*. 2019. doi: 10.1016/j.spinee.2018.07.005. **18.** Copay AG, et al. *Spine J*. 2008. doi: 10.1016/j.spinee.2007.11.006. **19.** Carlson et al. *Otolaryngol Head Neck Surg*. 2015. doi: 10.3171/2014.11.JNS14594/. **20.** Swigris et al. *Respir Med*. 2010. doi: 10.1016/j.rmed.2009.09.006.

Acknowledgments: The authors thank Nicole Coolbaugh of The Medicine Group (New Hope, PA, USA) for providing medical writing support, which was funded by Alder BioPharmaceuticals, Inc. (Bothell, WA, USA) in accordance with Good Publication Practice guidelines.

Disclosures: S. Tepper: *Consultant and/or scientific advisor:* Acorda, Alder BioPharmaceuticals, Alexsa, Allergan, Alphasights, Amgen, ATI, Axsome Therapeutics, Cefaly, Charleston Labs, DeepBench, Dr. Reddy's, ElectroCore, Eli Lilly, eNeura, GLG, Guidepoint Global, Magellan Rx Management, Neurolief, Nordic BioTech, Pfizer, Scion Neurostim, Slingshot Insights, Supernus, Teva, Zosano; *Editor and/or advising editor:* American Headache Society, Headache Currents, Headache, Wiley Blackwell; *Research support:* Alder BioPharmaceuticals, Allergan, Amgen, ATI, Dr. Reddy's Laboratories, ElectroCore, eNeura, Scion Neurostim, Teva, Zosano.: **D. Kudrow:** *Consultant and/or scientific advisor:* Alder BioPharmaceuticals, Amgen, Biohaven, Eli Lilly, Promius; *Research support:* Alder BioPharmaceuticals, Allergan, Amgen, Biohaven, Eli Lilly, Genentech, Roche, Teva, Viromed.: **R. Horblyuk:** *Contracted service provider:* Alder BioPharmaceuticals. **R. Cady:** *Full-time employee:* Alder BioPharmaceuticals; *Stockholder:* Alder BioPharmaceuticals. **E. Kassel:** *Full-time employee:* Alder BioPharmaceuticals; *Stockholder:* Alder BioPharmaceuticals. **J. Hirman:** *Contracted service provider:* Alder BioPharmaceuticals.

This study was funded by Alder BioPharmaceuticals, Inc.